EUFEPS - SITELF meeting, Naples 2026: Advances in pharmaceutical sciences

Transdermal Ointment in Cats: A Promising Strategy for Improved Compliance

E. Dauphin-Chanard, D. Pélisson
Gattefossé SAS, 69804 Saint-Priest, France

INTRODUCTION

Transdermal delivery represents an effective alternative to oral administration in cats, for which stress and handling constraints significantly reduce compliance. Mirataz® has demonstrated that the feline pinna allows efficient drug absorption and a prolonged half-life, enabling reduced dosing frequency while improving ease of administration and animal welfare[1,2].

The objective of this study was to develop a placebo ointment (DPE 268) based on Transcutol® V and Labrasol®, designed to mimic the physicochemical and rheological properties of Mirataz®. This formulation aims to serve as a robust platform for future incorporation of active pharmaceutical ingredients (APIs), supporting low-frequency transdermal dosing and improved treatment adherence.

MATERIALS & METHODS

Materials
Mirataz® 20 mg/g (Dechra) was used as the reference product. It contains macrogol 400, macrogol 3350, diethylene glycol monoethyl ether, caprylocaproyl polyoxyglycerides, oleyl alcohol, butylhydroxytoluene (E321), dimethicone and tapioca starch polymethyl silsesquioxane. DPE 268 formula is detailed in Table 1.

Methods
Ointments were prepared by heating excipients (except solubilizers) up to 75–80°C under controlled stirring (~ 500 rpm), followed by progressive cooling. Solubilizers were incorporated at 35°C under high shear (1000 rpm) for 10 minutes to prevent thickener crystallization. Finally, the stirring speed was reduced to 500 rpm and maintained until complete cooling.
Formulations were characterized using complementary approaches:
- Macroscopic evaluation, to assess appearance, color, and consistency;
- Microscopy, to assess microstructure homogeneity;
- Rheology (Haake Mars IQ Air rheometer, using the Haake RheoWin Data Manager software): oscillatory (amplitude sweep), steady-state (flow curve), and dynamic (thixotropy) tests were performed to evaluate structural organization, flow behavior, and structural recovery under shear.

Table 1. Composition of DPE 268 ointment formulation (% w/w) * Supplied by Gattefossé.

Excipients	Chemical name	Functionality	% (w/w)
Gelot™ 64*	Glyceryl monostearate NF and PEG-75 stearate NF	Non-ionic O/W emulsifier	12.0
Labrafac™ Lipophile WL 1349*	Medium-chain triglycerides NF	Oily phase	42.0
Geleol™ Mono & Diglycerides NF*	Glyceryl monostearate	Thickening agent	4.0
Glycerin	Propane-1,2,3-triol	Humectant	15.0
Transcutol® V*	Diethylene glycol monoethyl ether NF	Solubilizer	19.0
Labrasol®*	Caprylocaproyl macrogol-8 glycerides NF	Solubilizer	8.0

RESULTS

Both formulations exhibited a homogeneous white semi-solid appearance, with no visible phase separation or crystallization (Table 2). Microscopy confirmed a uniformly distributed microstructure, indicating good formulation stability.

Table 2. Characterization of Mirataz® vs DPE 268.

	Macroscopic appearance	Microscopic structure (×20 magnification)
Mirataz®	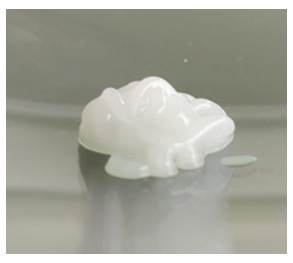	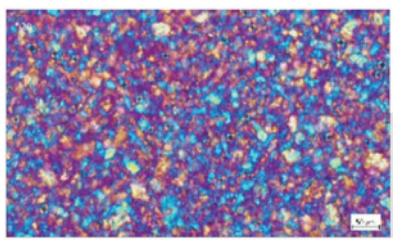
DPE 268	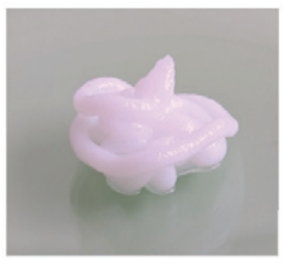	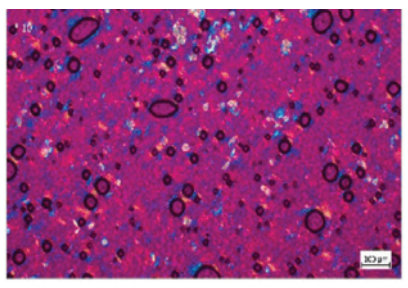

Rheological analysis showed that both systems behaved as structured viscoelastic networks, characteristic of ointment-type delivery systems. Amplitude sweep confirmed a gel-like behavior (G' > G'') for both formulations (Figure 1). DPE 268 exhibited higher elastic modulus values (G' ≈ 10^4 Pa) compared to Mirataz® (≈ 10^3 Pa), indicating a more rigid and highly structured network. In contrast, Mirataz® displayed a lower elastic modulus but greater resistance to progressive deformation, maybe due to interactions between the API and excipients affecting network organization.

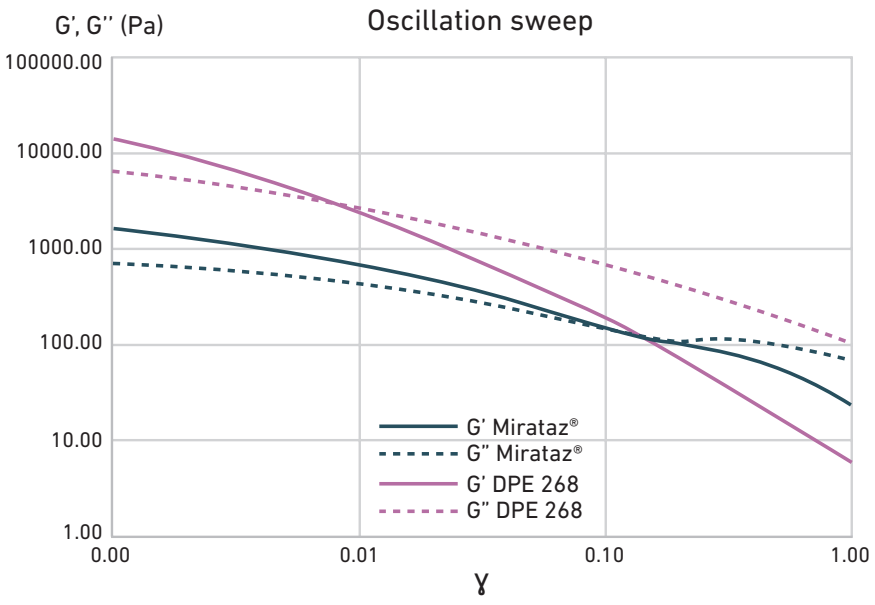
G', G'' (Pa)
Oscillation sweep
100000.00
10000.00
1000.00
100.00
10.00
1.00
0.00
0.01
0.10
1.00
γ
G' Mirataz®
G'' Mirataz®
G' DPE 268
G'' DPE 268
Figure 1. Comparison of network structure via amplitude sweep.

Flow curve analysis showed similar viscosities at low shear rates (≈ 10 – 100 Pa.s), confirming similar resting structure (Figure 2). At higher shear rates, Mirataz® reached ≈ 250 – 300 Pa.s, whereas DPE 268 remained lower (≈ 150 – 170 Pa.s), indicating increased resistance to flow, potentially due to API-induced intermolecular interactions or increased internal friction.

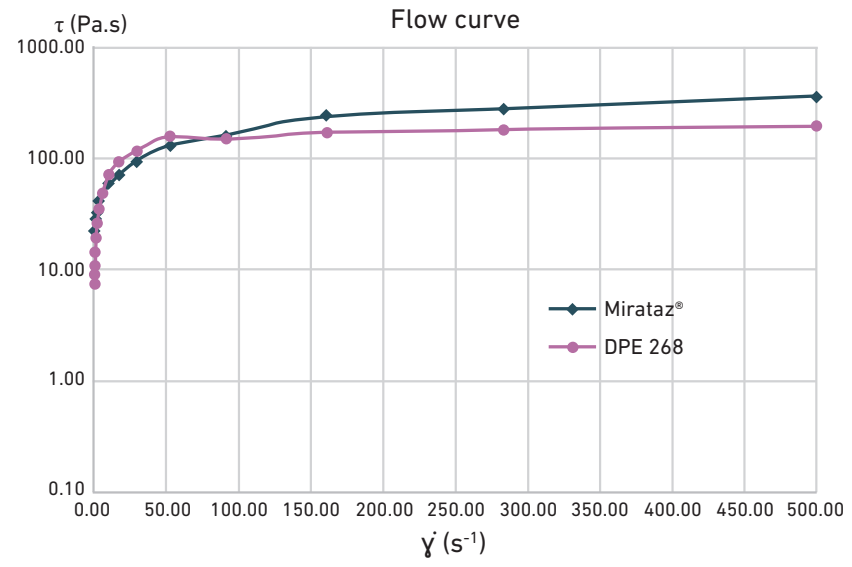
τ (Pa.s)
Flow curve
1000.00
100.00
10.00
1.00
0.10
0.00
50.00
100.00
150.00
200.00
250.00
300.00
350.00
400.00
450.00
500.00
γ̇ (s⁻¹)
Mirataz®
DPE 268
Figure 2. Shear-thinning behavior of Mirataz® and DPE 268.

Thixotropy tests revealed shear-dependent structural behavior for both formulations (Figure 3). DPE 268 exhibited a larger hysteresis loop (≈ 200 Pa versus ≈ 90 Pa at 20 s^{-1}), indicating greater structural breakdown under shear. Conversely, Mirataz® demonstrated faster structural recovery, reflecting a more resilient network, potentially influenced by reduced molecular mobility due to the API.

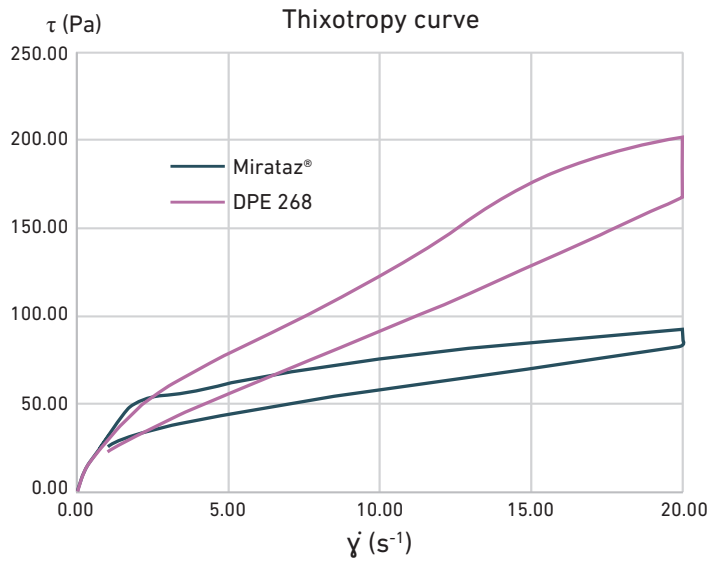
τ (Pa)
Thixotropy curve
250.00
200.00
150.00
100.00
50.00
0.00
0.00
5.00
10.00
15.00
20.00
γ̇ (s⁻¹)
Mirataz®
DPE 268
Figure 3. Structural breakdown and recovery: Thixotropy analysis.

Overall, both formulations displayed similar rheological signatures (viscoelastic, shear-thinning, thixotropic), supporting a Q3-like microstructural comparability. Importantly, flow curve results — being the most representative of application conditions — confirmed that both systems behaved similarly during use, while observed quantitative differences remain attributable to the presence of the active ingredient.

CONCLUSION

Despite moderate quantitative differences, the overall microstructural comparability demonstrates the robustness of the formulation approach, with variations consistent with active substance incorporation effects. **DPE 268 successfully reproduces the key physical and rheological characteristics of Mirataz®**, validating its use as a **transdermal placebo matrix**. The combination of **Transcutol® V and Labrasol® enhances skin penetration**, addressing a major limitation in feline therapy in which stress and oral dosing compromise compliance. By enabling safer and easier administration for both animals and caregivers, this formulation aligns with **One Health principles**, highlighting the interconnected benefits of improved animal welfare and human well-being[3].

REFERENCES

[1] https://medicines.health.europa.eu/veterinary/en/600000003654

[2] FABAD J. Pharm. Sci., 50, 3, 671-688, 2025

[3] C.M. Manasa, Journal of Drug Delivery Science and Technology 101 (2024) 106226